TIME TO WOUND CLOSURE AFTER AUTOGRAFT OR PLACENTAL-DERIVED ALLOGRAFT IN VLUs

POST HOC ANALYSIS OF CHRONEX

Snyder, R.J[1]; Haviv, A[1]; Singer, A.J[2]

1. MediWound Ltd 2. Renaissance School of Medicine at Stony Brook University, NY

MediWound

Overview

INTRODUCTION

Effective wound bed preparation and timely wound closure are essential in managing chronic lower-extremity ulcers. Evidence describing healing trajectories after autograft or allograft use in Venous Leg Ulcers (VLU) is limited. This post hoc analysis assessed closure incidence and timing following grafting procedures in the ChronEx randomized controlled trial.

METHODS

ChronEx was a multicenter, randomized, placebo-controlled study across 20 sites in the United States, Switzerland, and Israel. Patients with venous leg ulcers (VLUs) were randomized 3:3:2 to once-daily Bromelain-based Debridement (BBD), placebo gel vehicle (PLC), or non-surgical standard of care (NSSOC), for up to eight applications over 14 days or until complete debridement, followed by 12 weeks of non-active dressings. Autograft or allograft use during follow-up was permitted per investigator judgement, after complete debridement together with full coverage of the wound bed with granulation tissue has been achieved (wound bed prepared).

Complete closure was defined as 100% re-epithelialization without drainage or dressing need, confirmed at two weeks. Closure incidence and timing (mean, median, range) were summarized descriptively; no between-group statistical analyses were performed.

STUDY DESIGN

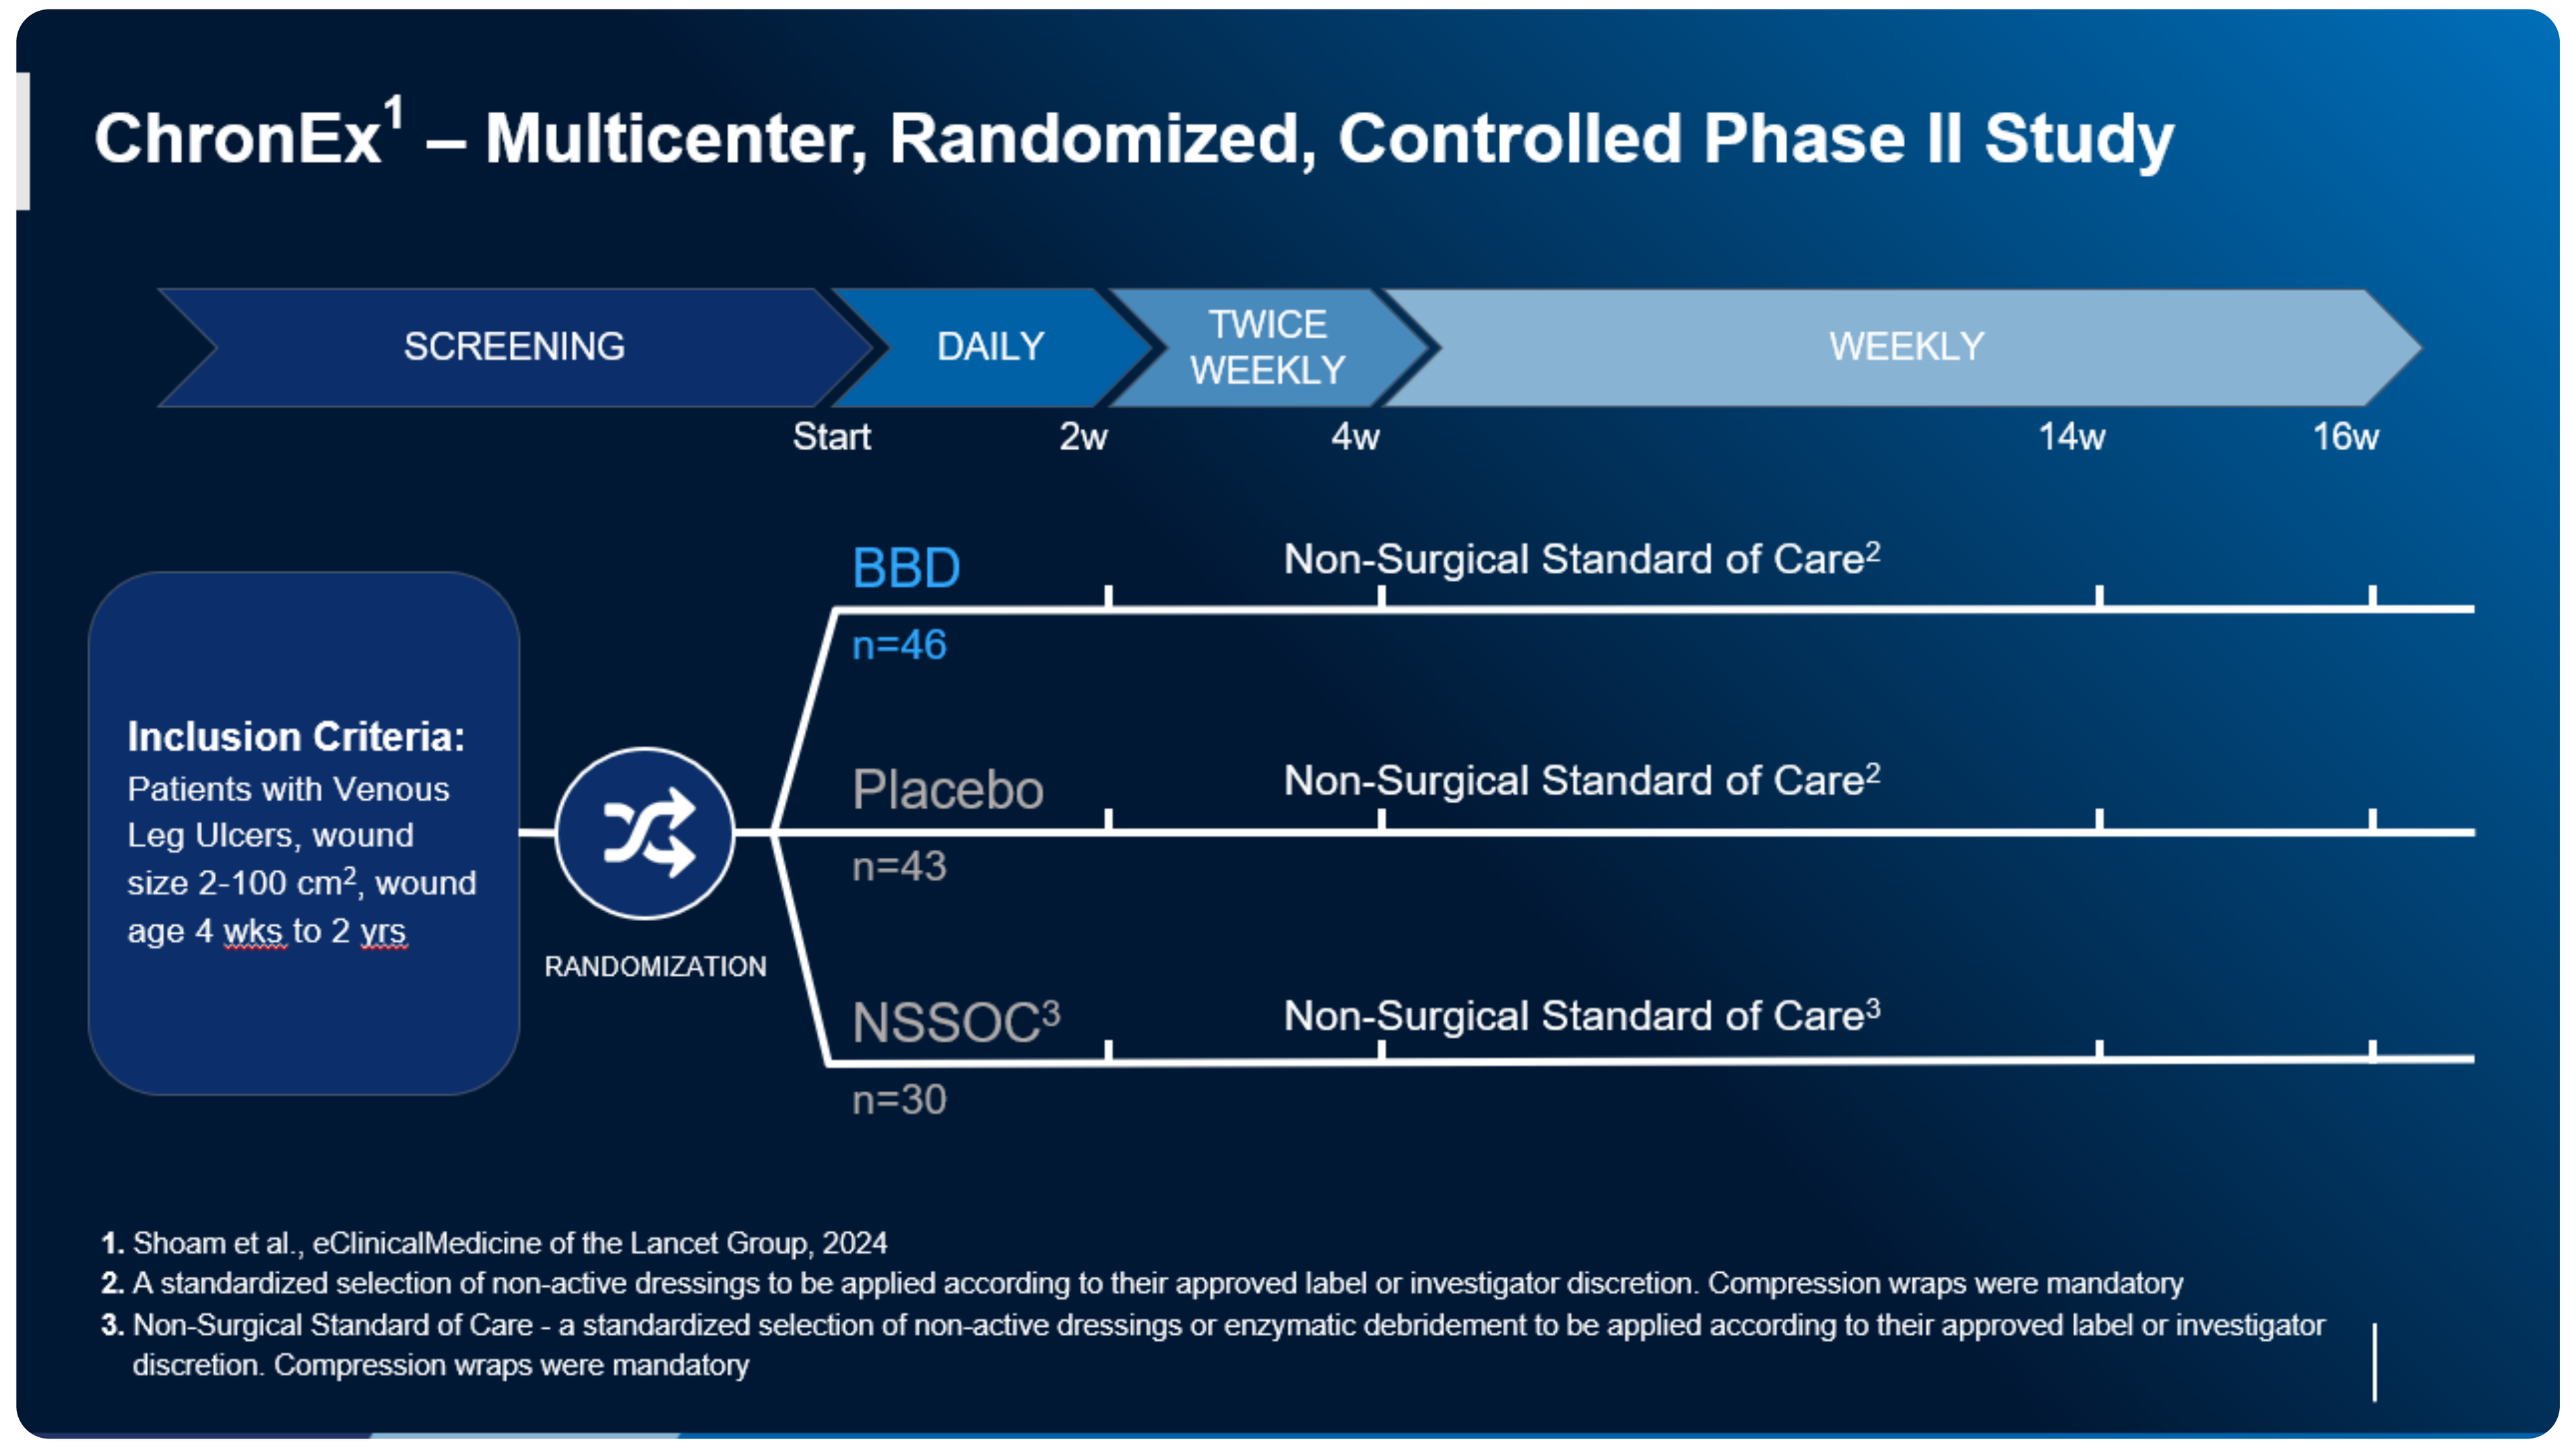
ChronEx[1] – Multicenter, Randomized, Controlled Phase II Study
SCREENING
DAILY
TWICE WEEKLY
WEEKLY
Start
2w
4w
14w
16w
Inclusion Criteria:
Patients with Venous Leg Ulcers, wound size 2-100 cm², wound age 4 wks to 2 yrs
RANDOMIZATION
BBD
n=46
Non-Surgical Standard of Care[2]
Placebo
n=43
Non-Surgical Standard of Care[2]
NSSOC[3]
n=30
Non-Surgical Standard of Care[3]
1. Shoam et al., eClinicalMedicine of the Lancet Group, 2024
2. A standardized selection of non-active dressings to be applied according to their approved label or investigator discretion. Compression wraps were mandatory
3. Non-Surgical Standard of Care - a standardized selection of non-active dressings or enzymatic debridement to be applied according to their approved label or investigator discretion. Compression wraps were mandatory

Shoham et al. 2024; LANCET's *eClinicalMedicine*

Results

- Nine patients received autografts(BBD n=5, PLC n=3, NSSOC n=1)
- Seven patients received placental-derived allografts (BBD n=2, PLC n=3, NSSOC n=2)

BASELINE CHARACTERISTICS

Parameter	Autograft (n=9)	Allograft (n=7)
Mean Age	68.2	71.4
Median Wound Size	5.0 cm²	5.4 cm²
Median Wound Age	36 weeks	19 weeks

HIGHER INCIDENCE OF WOUND CLOSURE IN AUTOGRAFTED WOUNDS

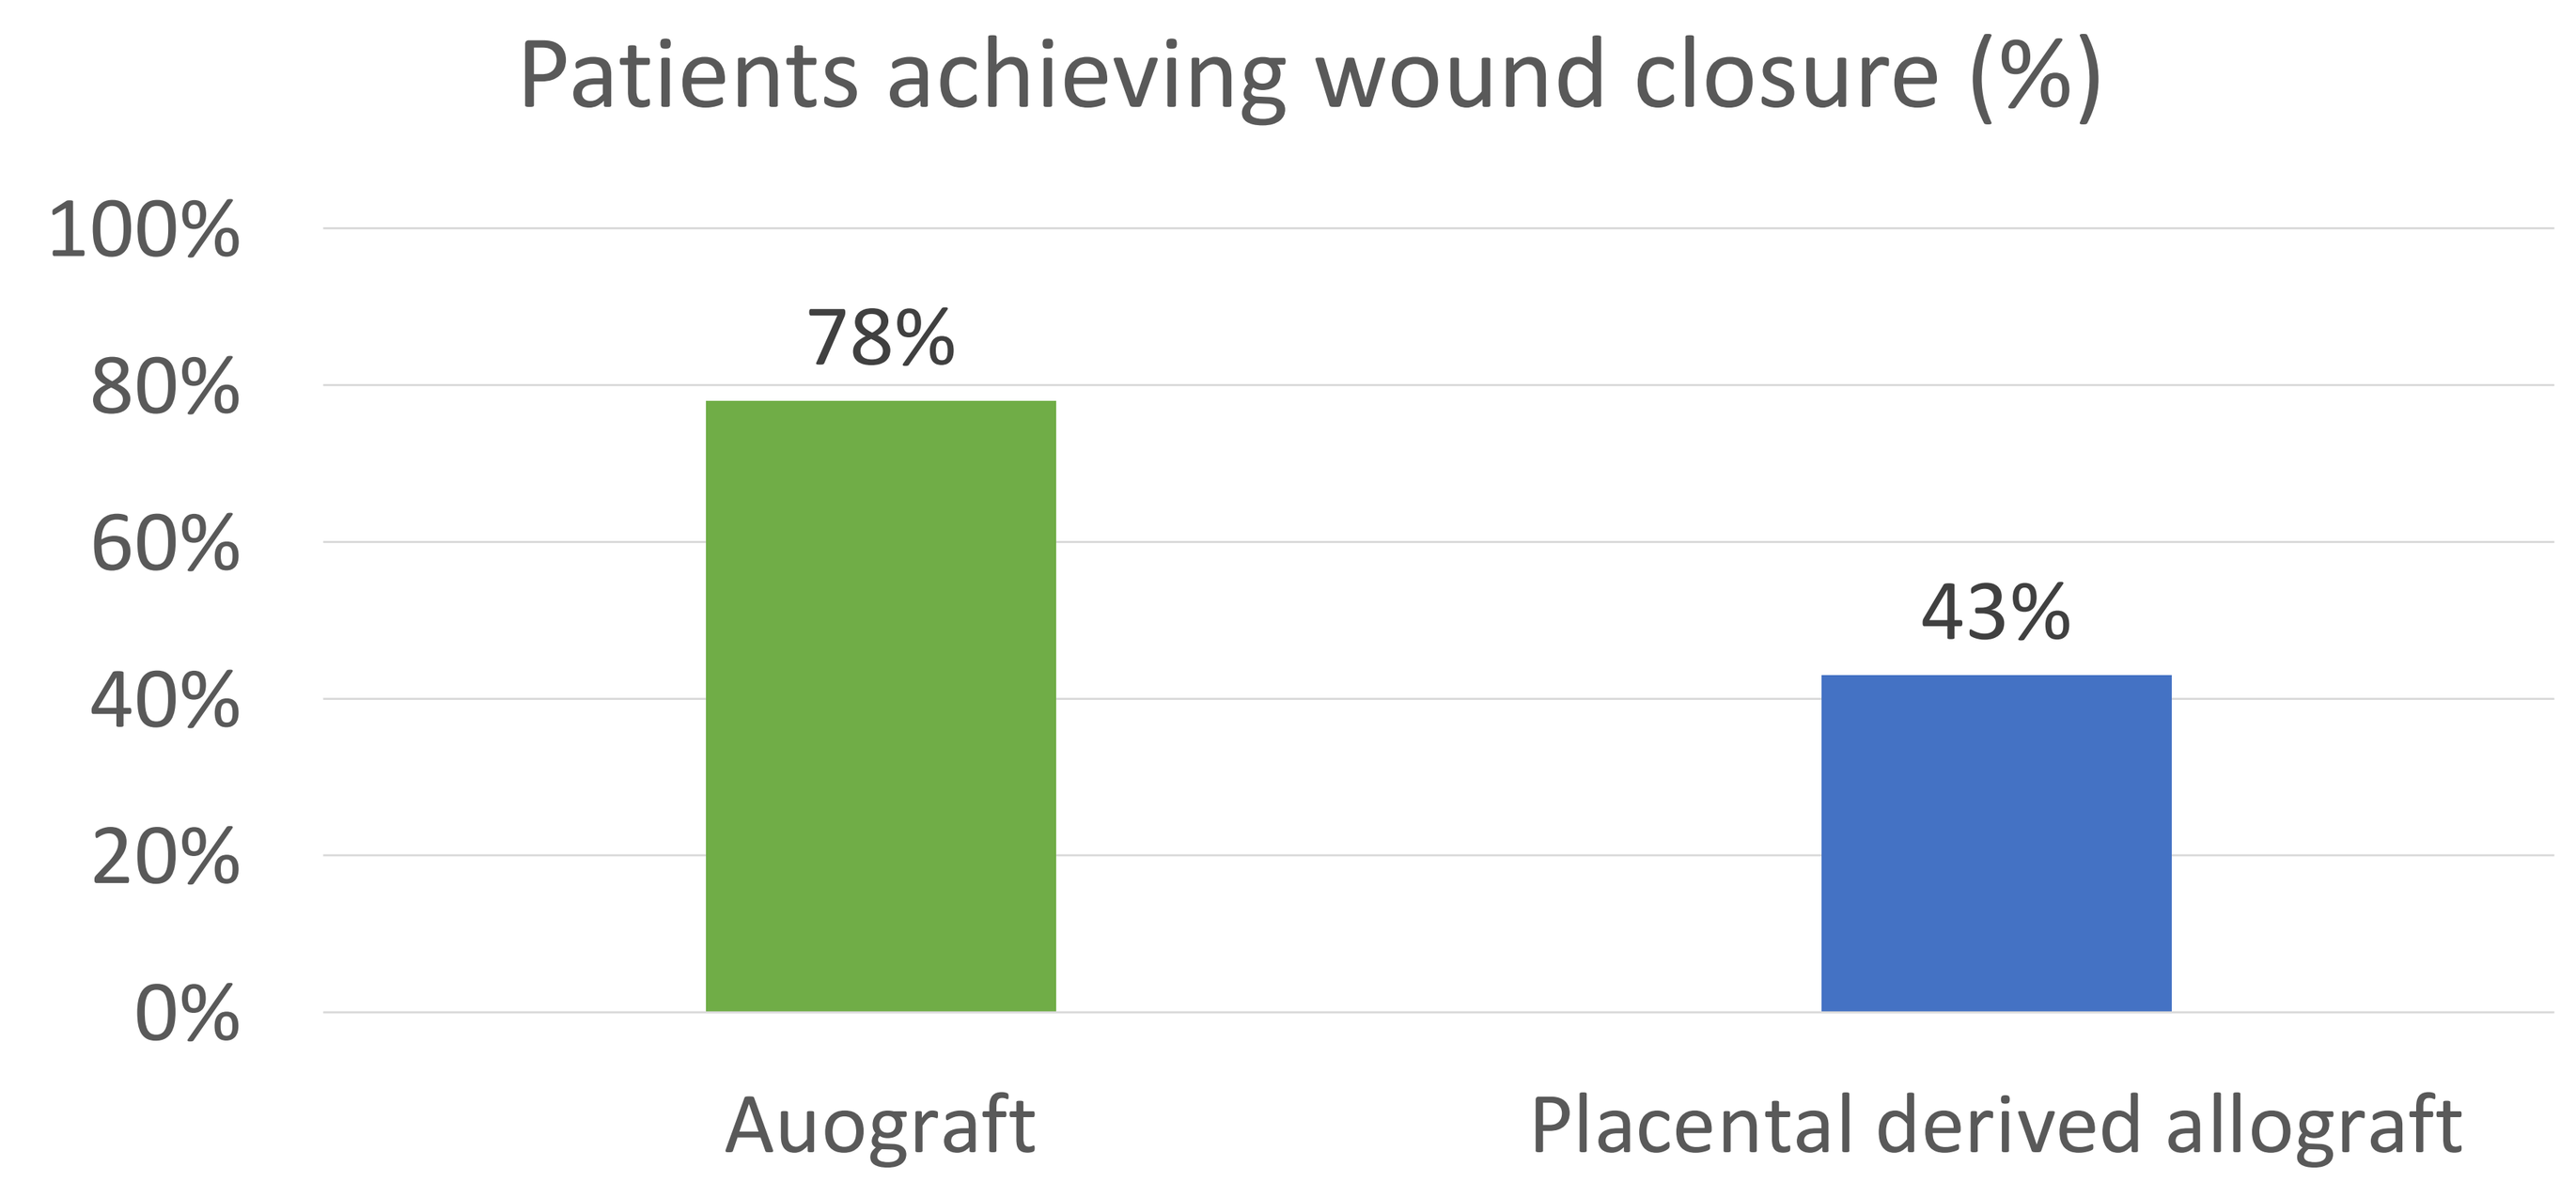
Patients achieving wound closure (%)
100%
80%
60%
40%
20%
0%
78%
43%
Auograft
Placental derived allograft

SHORET TIME TO WOUND CLOSURE IN AUTOGRAFTED WOUNDS

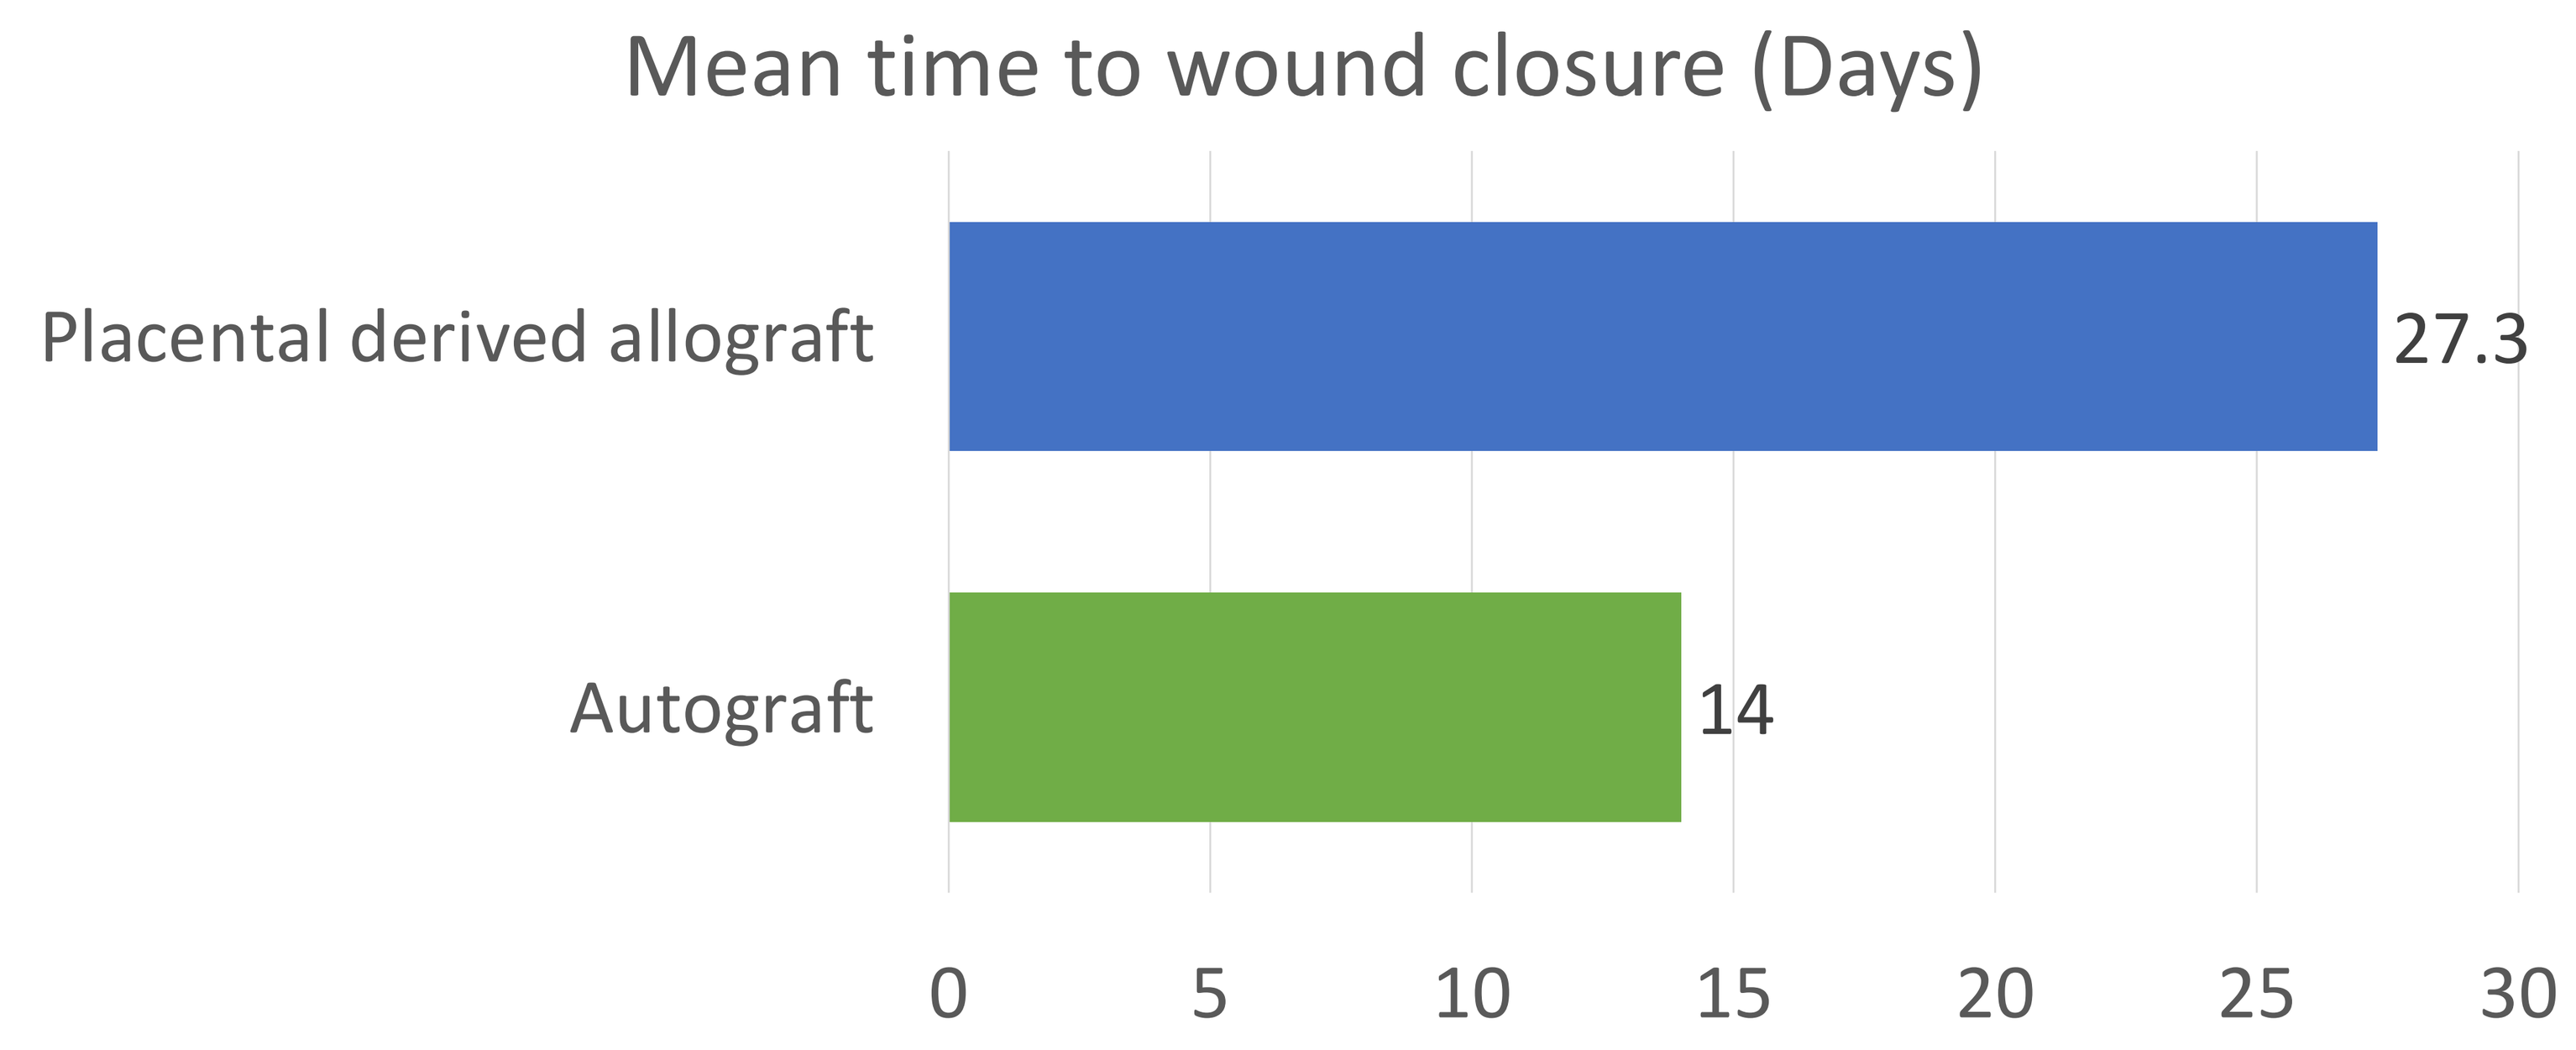
Mean time to wound closure (Days)
Placental derived allograft
27.3
Autograft
14
0
5
10
15
20
25
30

Conclusions

- Autografting after wound bed preparation yielded higher closure rates and faster healing (≈2–3 weeks), whereas allograft-treated wounds showed lower closure incidence and slower trajectories (≈3–6 weeks)
- These findings provide practical insight into expected healing timelines following grafting in VLU

- Data confirm the statistical assumptions of the ongoing global Phase III VALUE EscharEx® (BBD) study in VLU, based on the ChronEx RCT
- The VALUE trial mandates active wound closure with either autograft or placental-derived allograft (EpiFix®) following achievement of an adequate wound bed and includes time to wound closure as a co-primary endpoint