ESCHAREX PROMOTES RAPID DEBRIDEMENT AND MODULATES WOUND HEALING–RELATED GENE EXPRESSION IN A PORCINE CHRONIC WOUND MODEL

Sanders, M.[1]; Hanna, M.[1]; Angel, A.[2]; Geblinger, D.[2]
1. ProDevLabs, Southborough, Massachusetts, USA; **2.** MediWound Ltd;

MediWound

Overview

INTRODUCTION

Effective debridement is a critical prerequisite for timely wound healing. Collagenase-based enzymatic debridement is widely used but is often associated with gradual and slow non-viable tissue removal, which may delay wound closure. EscharEx® is a novel bromelain-based enzymatic debridement agent developed to accelerate the breakdown of non-viable tissue while promoting a wound environment conducive to healing.

In this study, we explored debridement efficacy and the molecular mode of action of EscharEx compared with placebo and SANTYL in a porcine chronic wound model.

METHODS

Twelve full-thickness hard-to-heal wounds were created on the dorsal side of 4 pigs by skin excision combined with doxorubicin injection on days 1 and 6 post-wounding. The wound bed was characterized by a distinct central slough and peripheral full-thickness necrotic skin. Twenty days after model establishment, wounds were treated with EscharEx, SANTYL, or gel vehicle (placebo control). Treatments were applied once daily for 8 days. Debridement progression was assessed using blinded clinical scoring, quantitative image analysis, and histological evaluation. Serial wound biopsies were collected at multiple time points and analyzed by RNA sequencing.

Debridement Results

In the chronic wound model, peripheral non-viable tissue was generated by intradermal injection of doxorubicin and consisted of full-thickness necrotic skin. In the center of the wound, all skin layers were removed, resulting in slough formation adherent to the wound bed. EscharEx removed both slough and full-thickness necrotic skin significantly more rapidly than placebo and SANTYL.

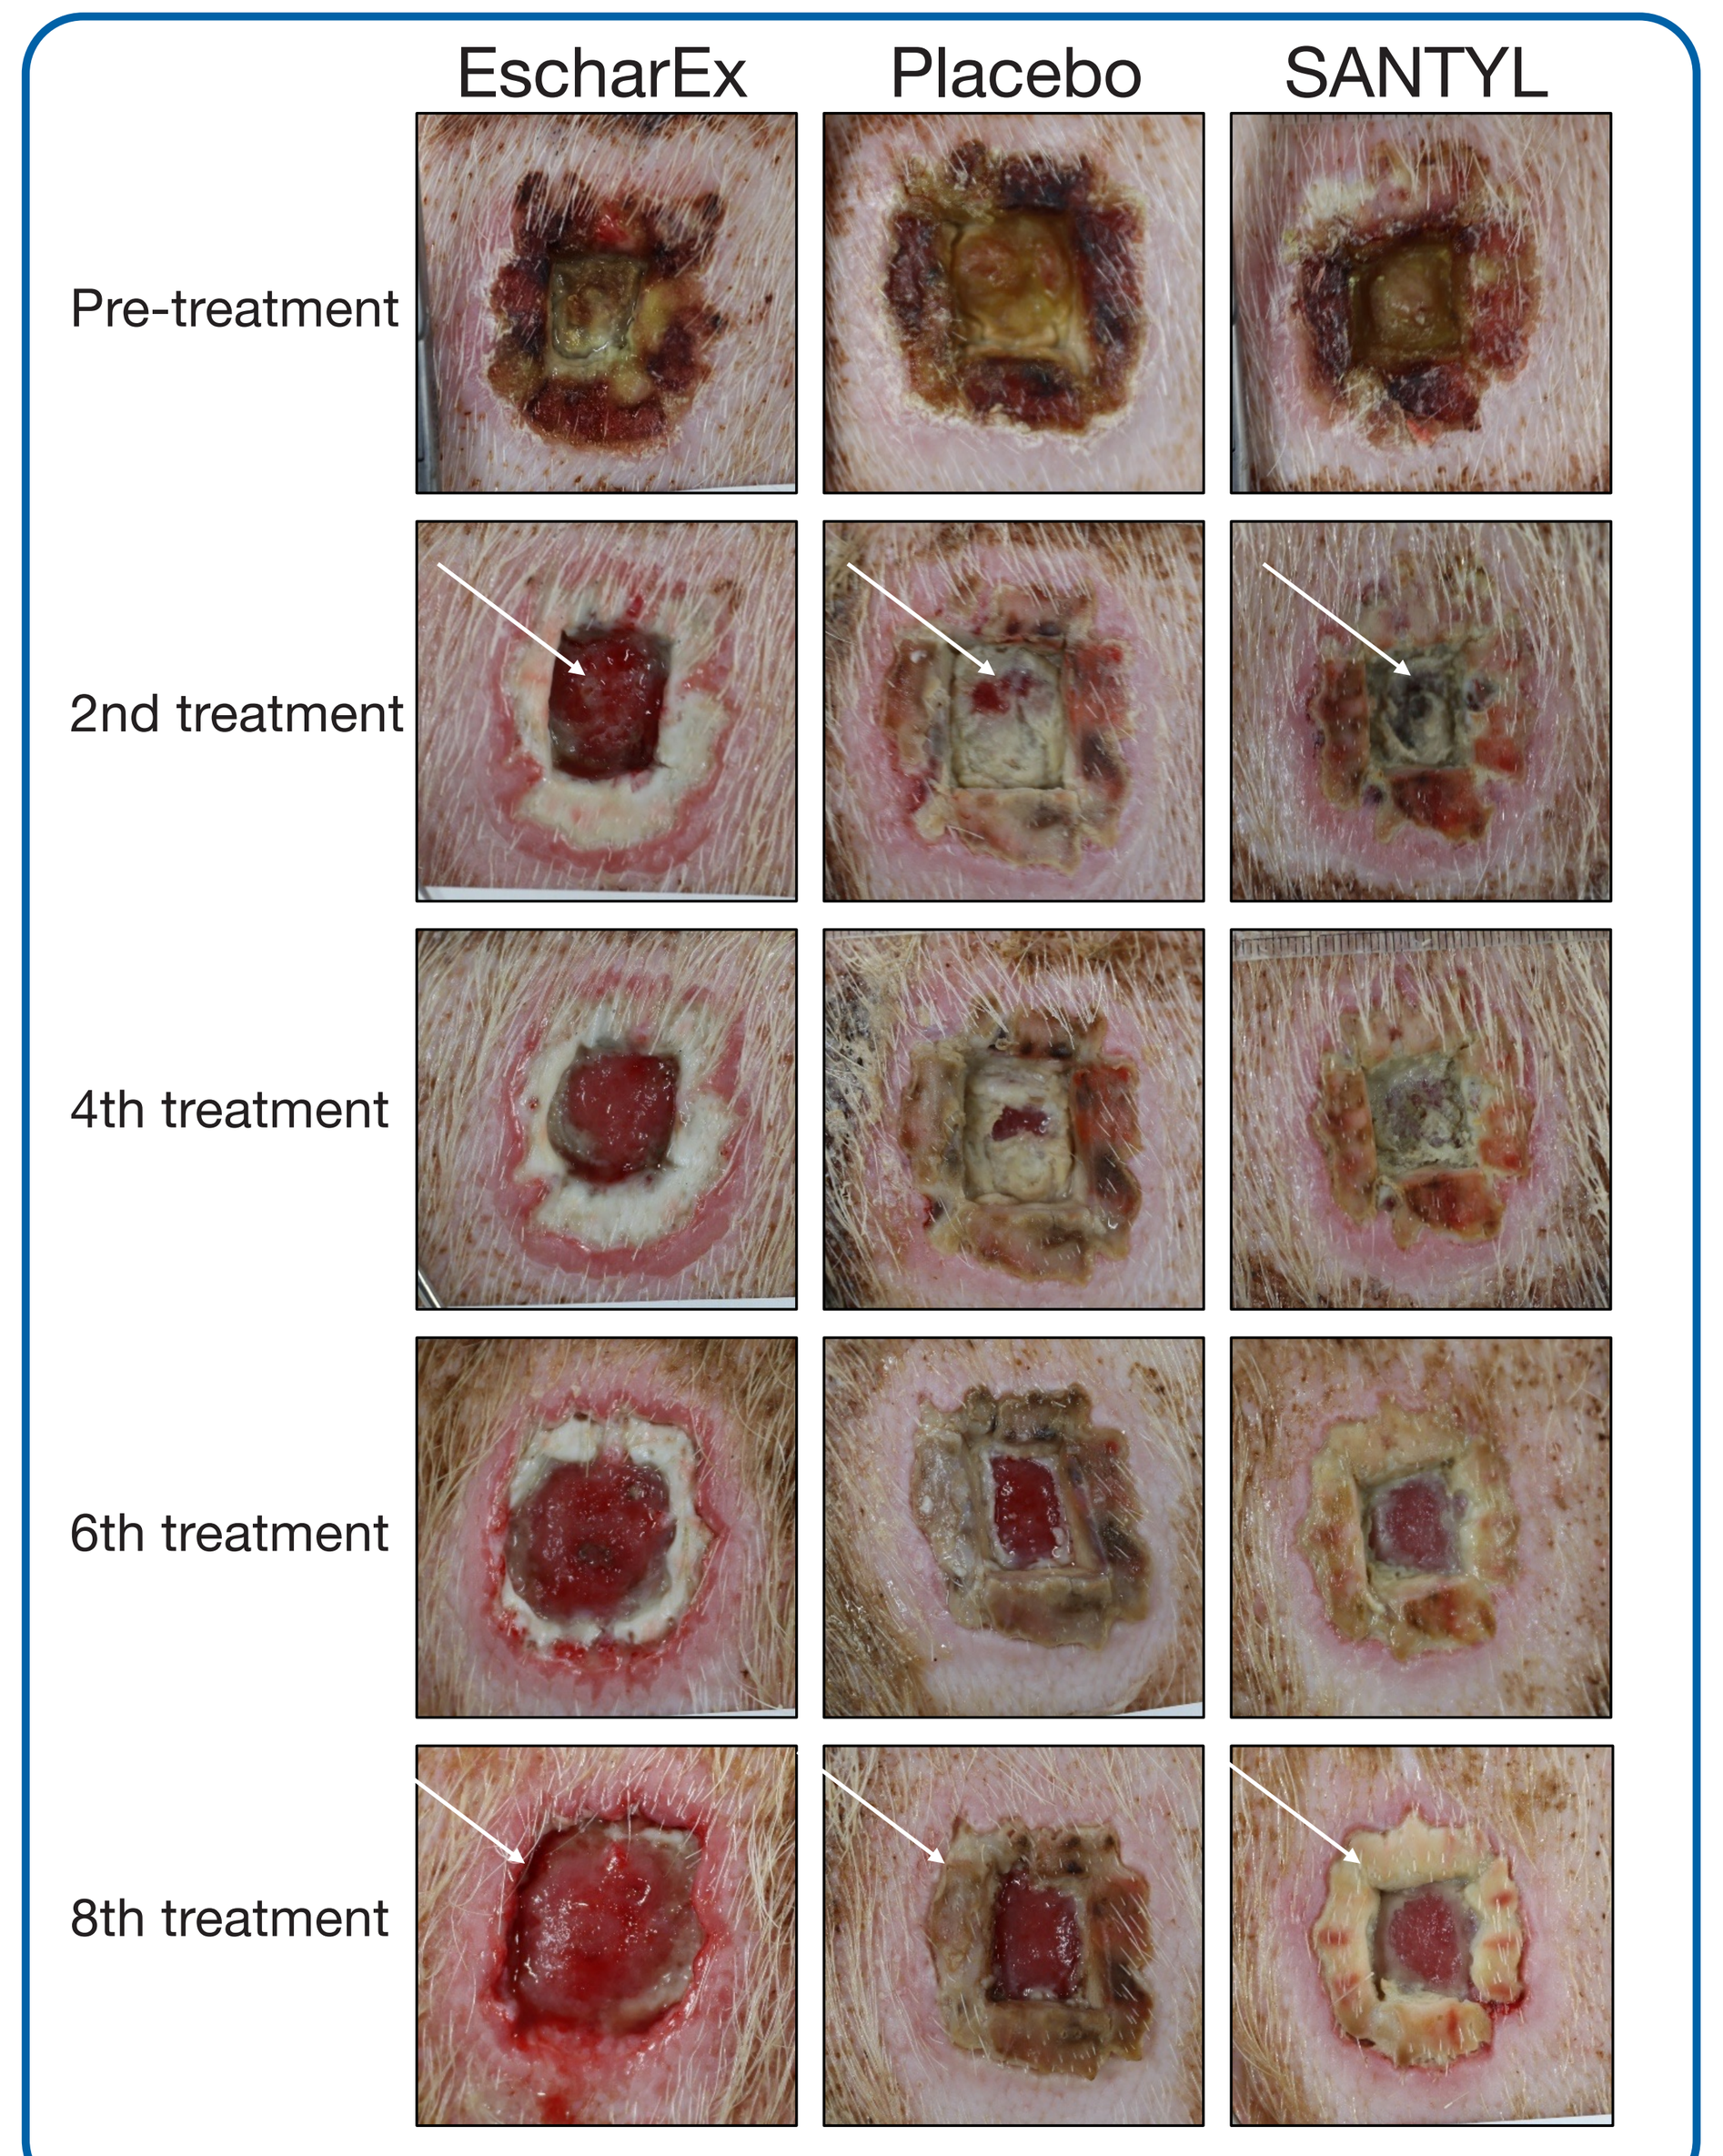
EscharEx
Placebo
SANTYL
Pre-treatment
2nd treatment
4th treatment
6th treatment
8th treatment

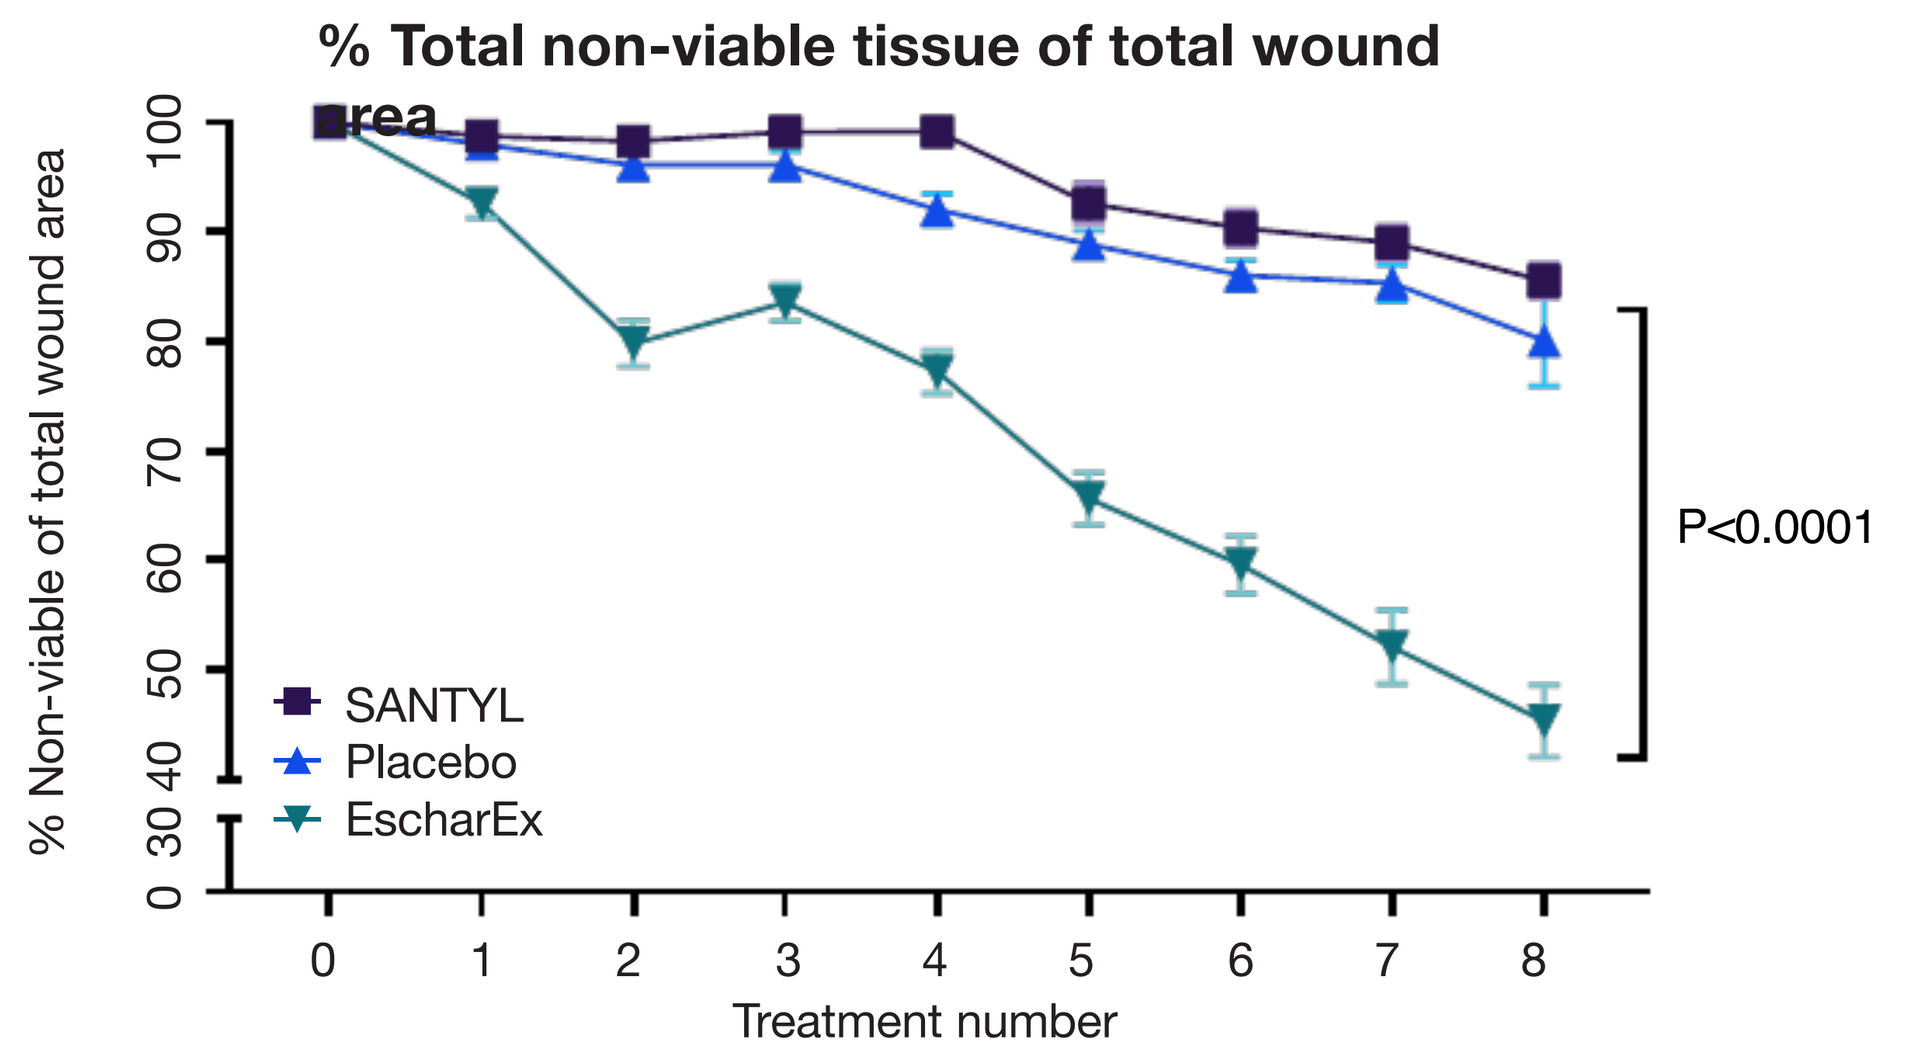
% Total non-viable tissue of total wound area
% Non-viable of total wound area
SANTYL
Placebo
EscharEx
P<0.0001
Treatment number

P<0.0001 from 2nd treatment

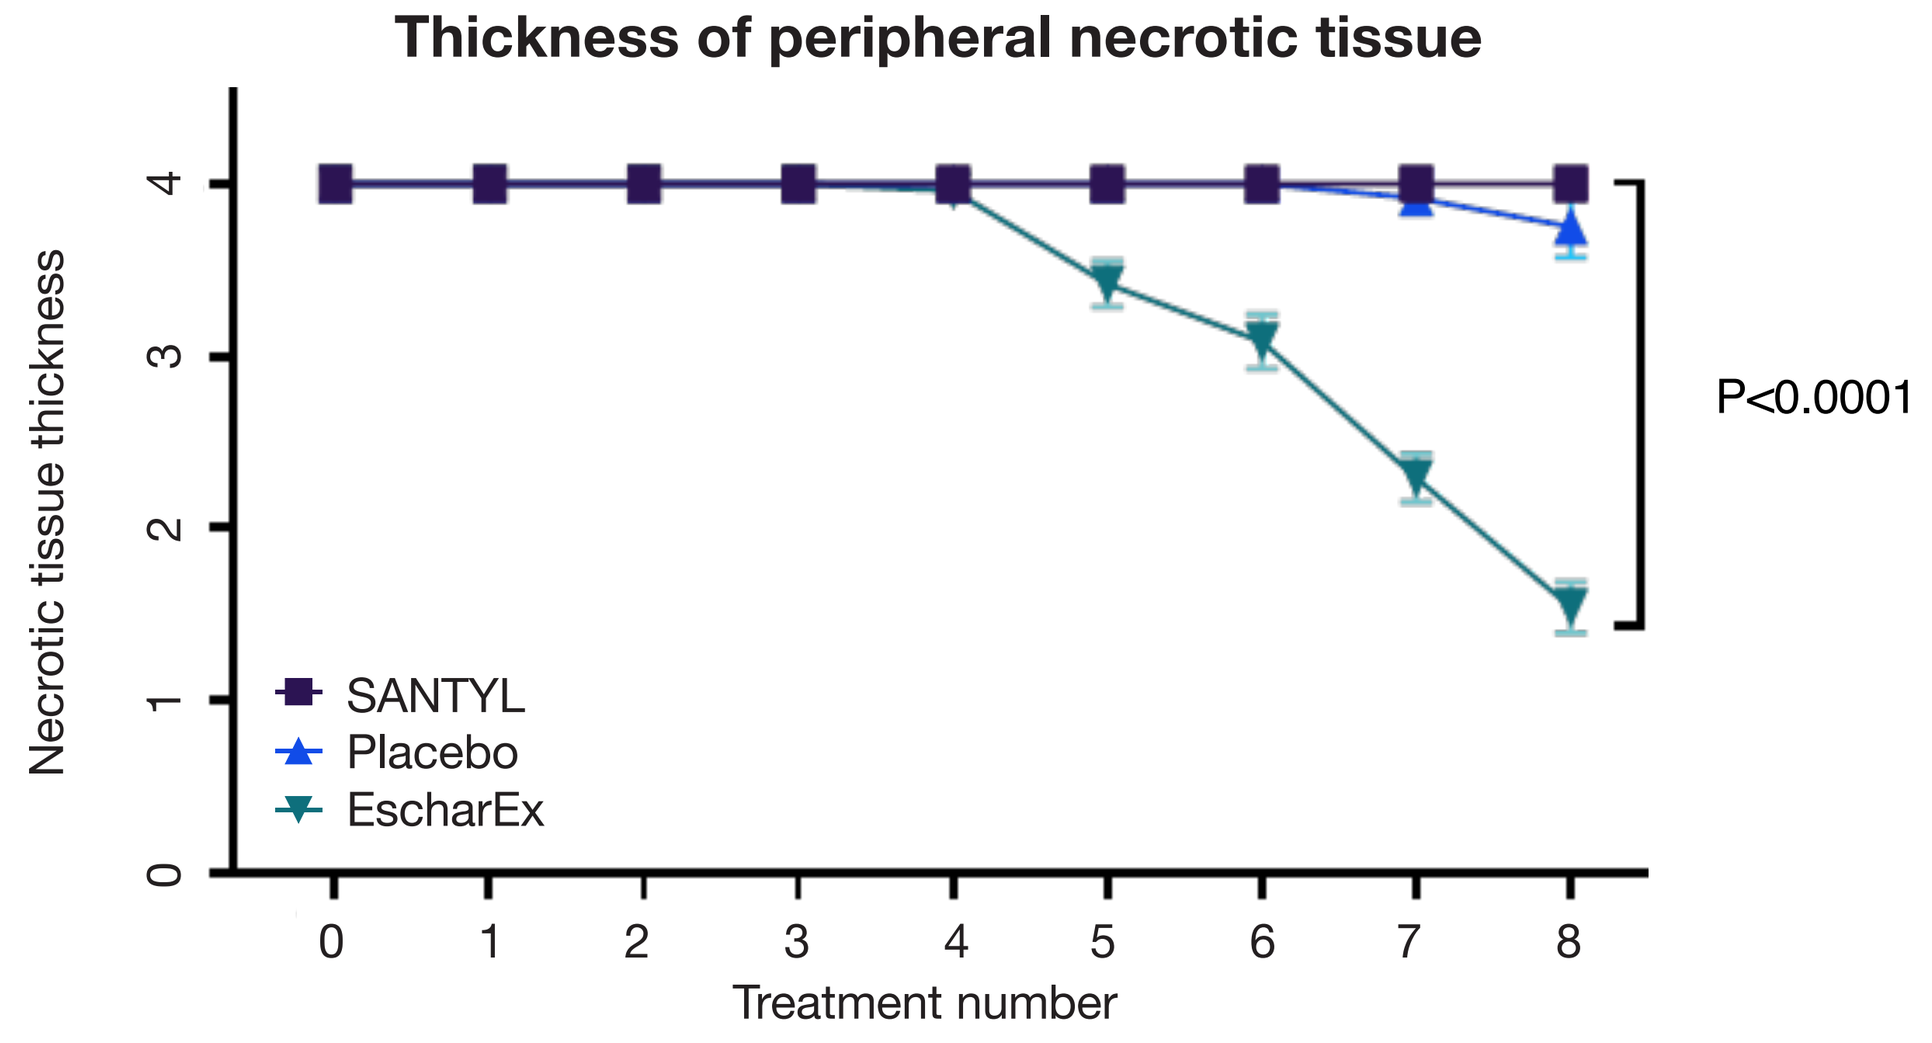
Thickness of peripheral necrotic tissue
Necrotic tissue thickness
SANTYL
Placebo
EscharEx
P<0.0001
Treatment number

P<0.001 from 5th treatment

Modulation of Wound Healing

VEGDF - Vascular Endothelial Growth Factor D

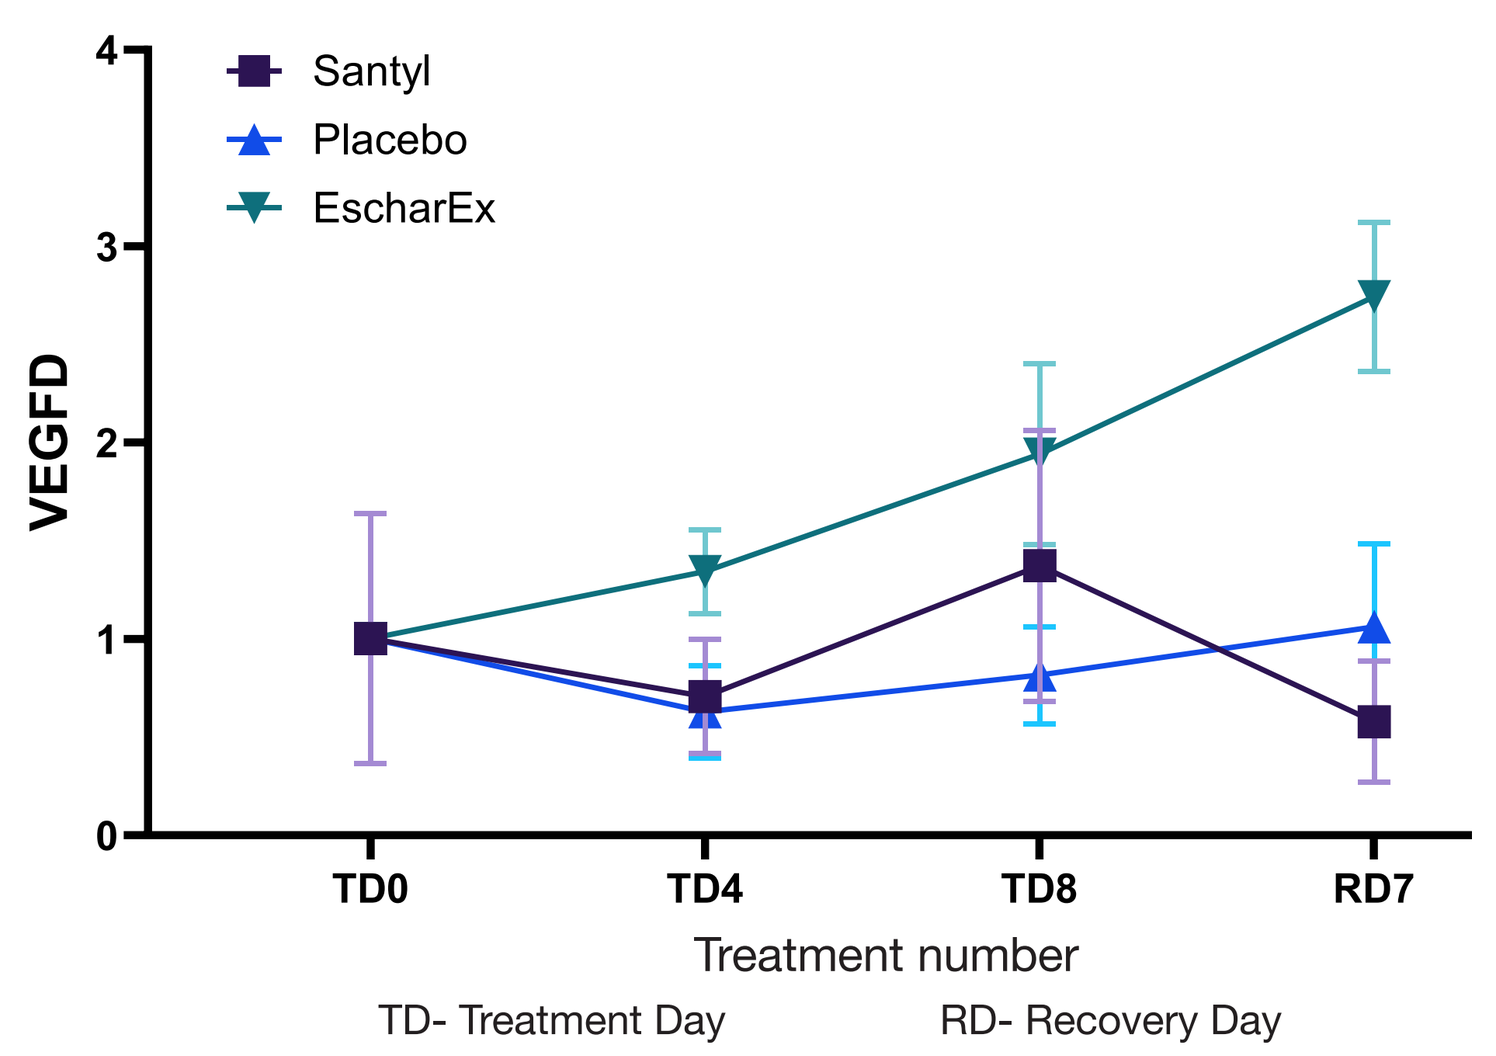
VEGFD
Santyl
Placebo
EscharEx
TD0
TD4
TD8
RD7
Treatment number
TD- Treatment Day
RD- Recovery Day

VEGFD promotes **angiogenesis** and stimulates **endothelial cell migration**; critical for establishing **vascular networks** supporting tissue **regeneration**

FGF7 (KGF) - Fibroblast Growth Factor 7

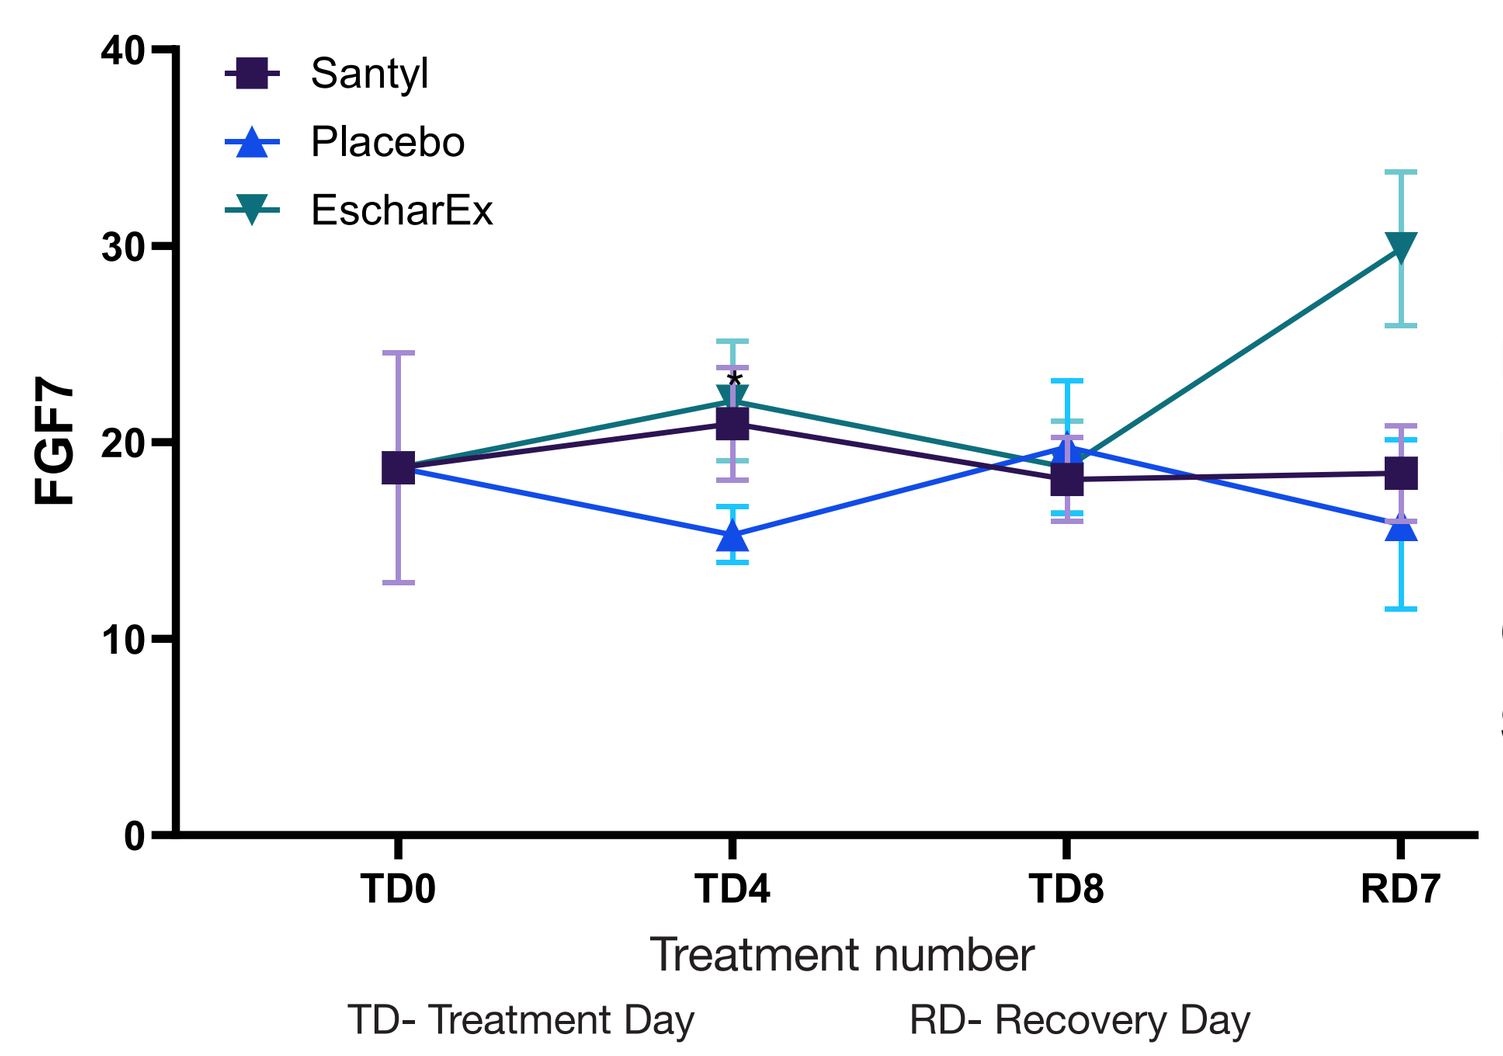
FGF7
Santyl
Placebo
EscharEx
TD0
TD4
TD8
RD7
Treatment number
TD- Treatment Day
RD- Recovery Day

FGF7 stimulate **keratinocyte** migration; enhance **re-epithelialization**; promotes **wound closure** and **reduce scarring**

CD59 - Protectin

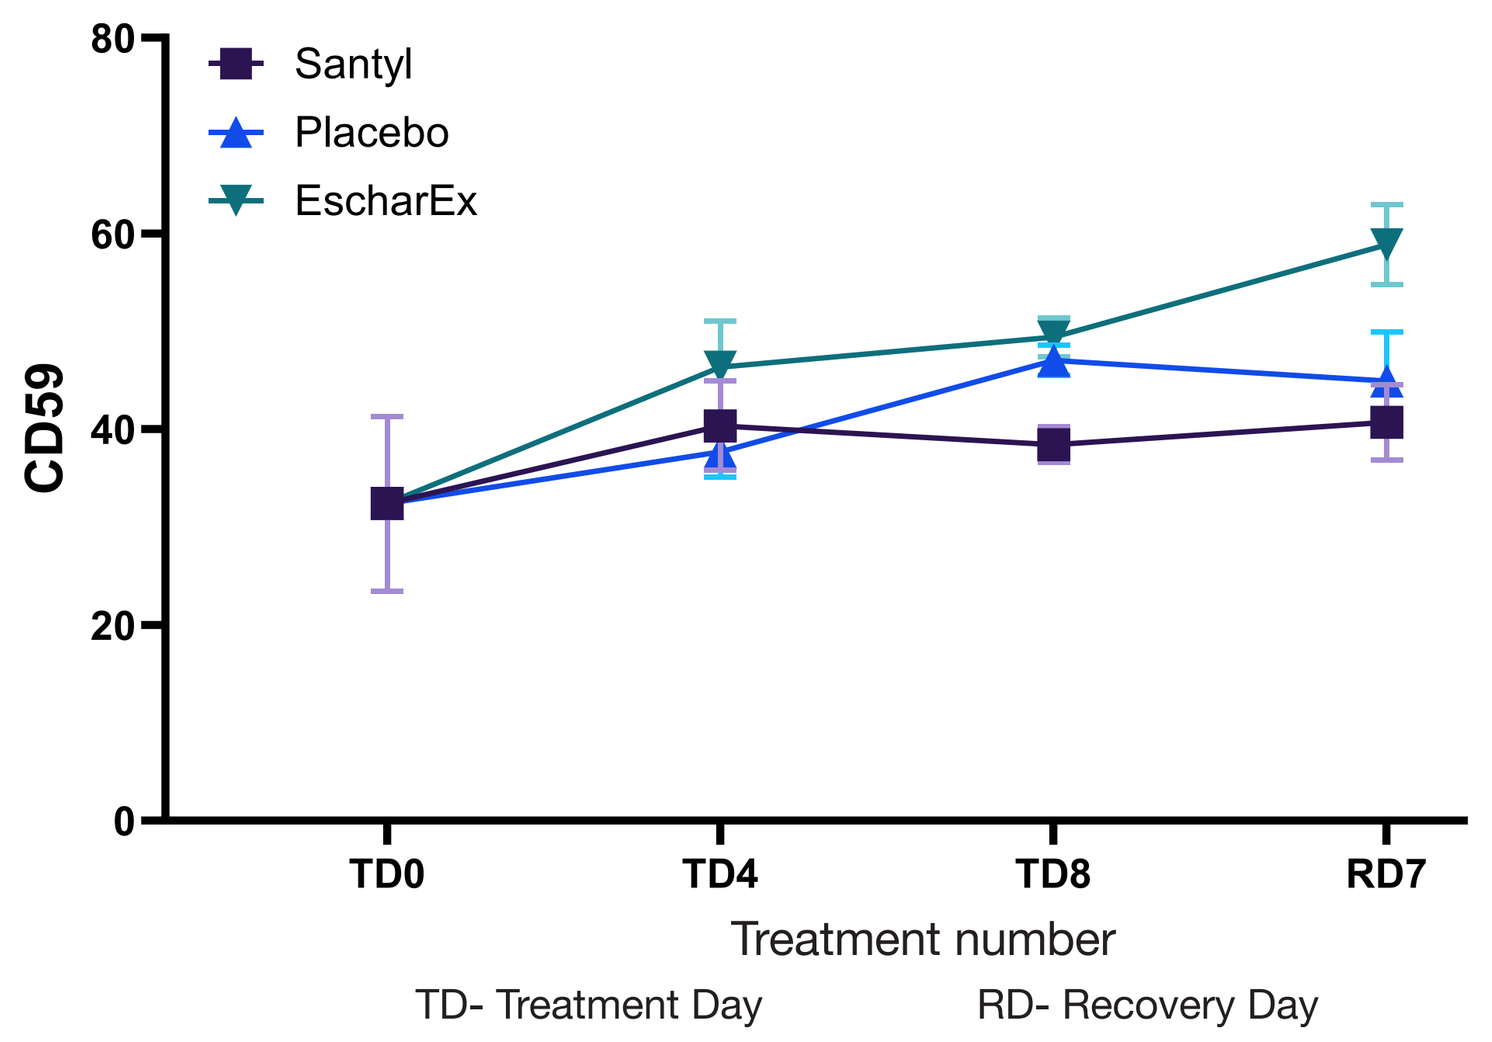
CD59
Santyl
Placebo
EscharEx
TD0
TD4
TD8
RD7
Treatment number
TD- Treatment Day
RD- Recovery Day

CD59 limits membrane attack complex (MAC) formation and **prevent necrosis**; up-regulation of CD59 **reduce skin inflammation.**

MMP13 - Matrix Metalloproteinase-13

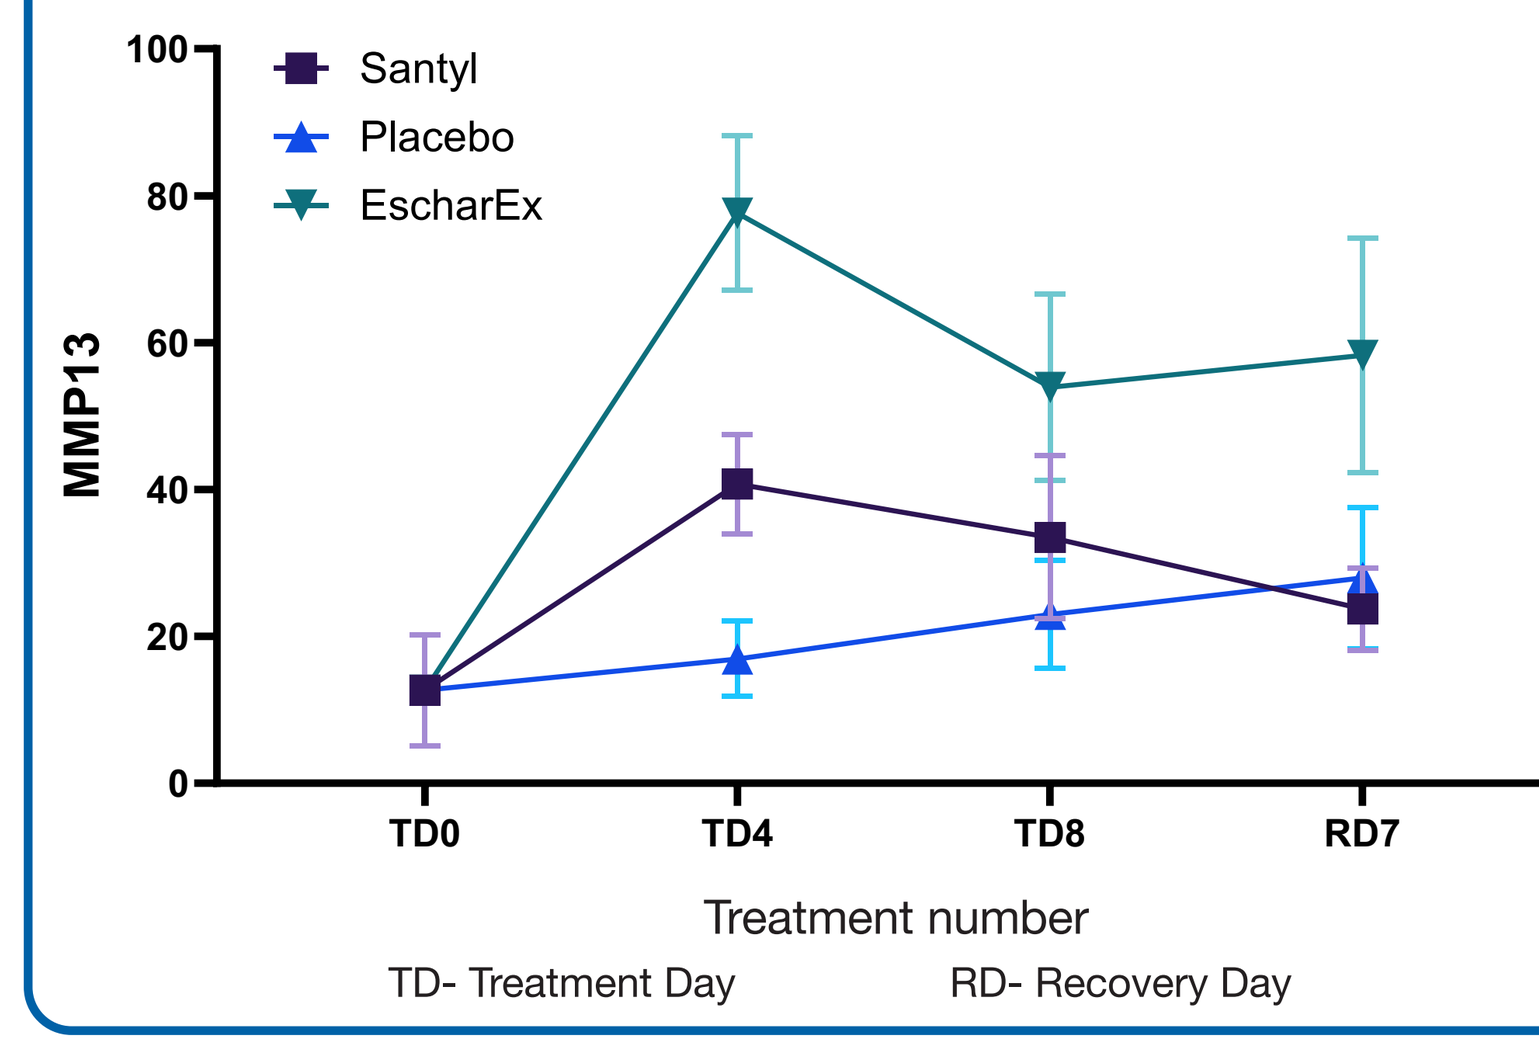
MMP13
Santyl
Placebo
EscharEx
TD0
TD4
TD8
RD7
Treatment number
TD- Treatment Day
RD- Recovery Day

MMP13 is expressed by keratinocytes; enhances 3D **collagen remodeling**; supports **fibroblast** survival; regulates **granulation tissue** growth

WISP1 - Inducible Signaling Pathway Protein 1

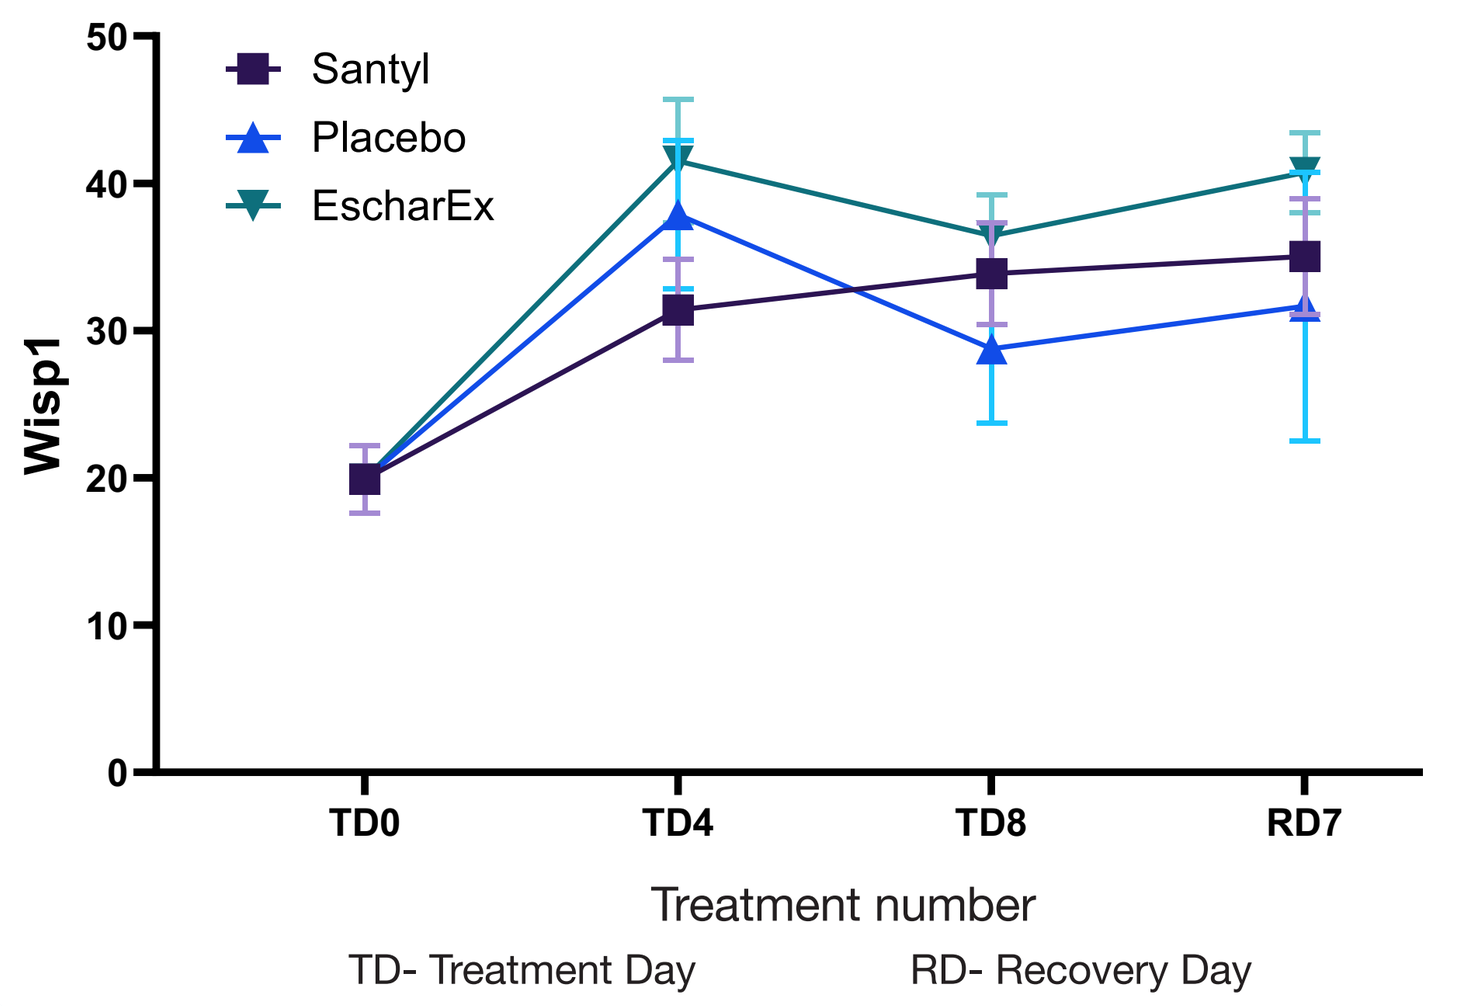
Wisp1
Santyl
Placebo
EscharEx
TD0
TD4
TD8
RD7
Treatment number
TD- Treatment Day
RD- Recovery Day

WISP1 (WNT1 / CCN4) stimulates dermal **fibroblast** migration; Promotes extracellular matrix **remodeling;** accelerates **wound closure**

LTA - Lymphotoxin Alpha

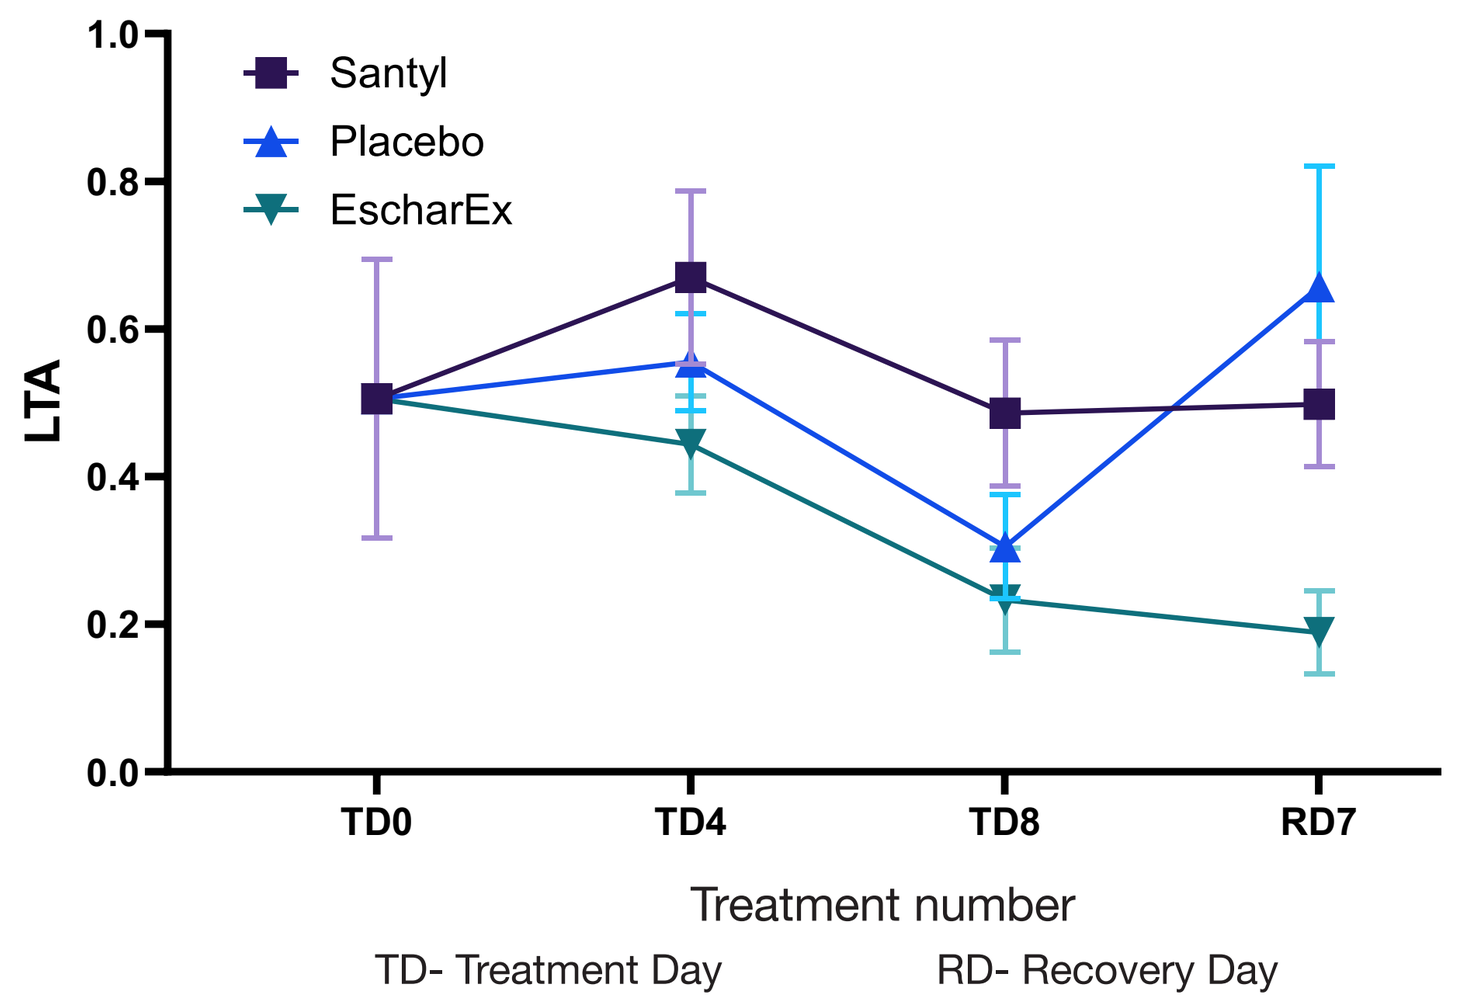
LTA
Santyl
Placebo
EscharEx
TD0
TD4
TD8
RD7
Treatment number
TD- Treatment Day
RD- Recovery Day

LTA is an inflammatory cytokine; promotes inflammatory responses; Declining LTA **avoids prolonged inflammation**

Conclusions

EscharEx demonstrated:

- **Superior debridement:** Effective removal of various types of non-viable tissue commonly found across different chronic wound etiologies, outperforming SANTYL and placebo
- **Modulation of key biological pathways supporting wound healing** evident by gene expression changes associated with:
 - **Necrosis prevention**
 - **Angiogenesis** and **granulation tissue** formation
 - **Migration and survival** of fibroblasts, keratinocytes and endothelial cells
 - **Modulation of inflammation**
 - **Remodeling** of collagen and extracellular matrix